Utility of ctDNA-based Targeted Methylation MRD Assay for Hematologic Malignancies

Veerendra Munugalavadla[1], Gary De Jesus[1], Aleksandra Markovets[2], Qinwen Liu[3], Oliver Venn[3], Rafael White[1], Giulia Fabbri[2], Paul Labrousse[2], Dan Stetson[2], Brian Dougherty[2], Darren Hodgson[4], Jill Walker[4], Anas Younes[5], Daniel Auclair[2]

[1]AstraZeneca, South San Francisco, CA, USA; [2]AstraZeneca, Waltham, MA, USA; [3]GRAIL, LLC, Menlo Park, CA, USA; [4]AstraZeneca, Cambridge, UK; [5]AstraZeneca, New York, NY, USA

3369

Introduction

- MRD is an emerging independent predictor of progression-free survival and overall survival in several hematologic diseases[1,2]
- Despite the use of many different MRD assays in clinical trials, the lack of standardization, heterogeneity of diseases, and need for primary tumor sample hinder the use of MRD assays for several blood cancers outside of the research setting[1,3]
- Methylated ctDNA approaches have emerged as promising techniques for early cancer detection[4]
 - DNA methylation patterns are altered in many hematologic malignancies[5,6]; therefore, methylated DNA may be a good marker for the detection of residual cancer DNA in the blood[5]
- This study assessed the feasibility of a pan hematologic malignancy classifier based on a novel methylated ctDNA-based platform developed by GRAIL, LLC (Menlo Park, CA, USA) as a potential tumor-agnostic, plasma-based MRD assay for hematologic malignancies

Methods

- The methylated ctDNA assay, developed by GRAIL, LLC, for MRD assessment uses blood-only liquid biopsy and estimates tumor burden using methylation signatures and predicts MRD status based on a pan-heme classifier
 - MVAF leverages the methylation patterns of cancer-derived DNA to estimate the variant allele fraction of a cell-free DNA sample[7]
 - MVAF is estimated based on the Bayesian statistical inference method; the GRAIL Bayesian inference generates a full posterior probability distribution of probability mass[7]
 - The credible interval of the MVAF is the central portion of the posterior distribution that contains 95% of the MVAF values[7]
- Plasma samples from patients with DLBCL, FL, MCL, AML, CLL, and MM were tested retrospectively using the novel GRAIL heme classifier
- Cancer signal detection and CSO prediction were explored at various detection specificity thresholds
 - A detection specificity threshold is a detection threshold that achieves a prespecified specificity level in a non-cancer population (a 98% detection specificity threshold is designed to achieve a test positive call rate of <2% in individuals without cancer)
- Reproducibility of cancer detection was assessed using paired samples that were taken prior to treatment at screening (T0) and cycle 1, day 1 (T1) ("double baseline samples")
- Sensitivity of the GRAIL heme classifier was evaluated in post-treatment samples using orthogonal methods
- LOD of the GRAIL heme classifier was estimated via analysis of contrived samples generated by spiking serially diluted DLBCL and CLL samples into healthy volunteer plasma samples
- Concordance was evaluated by comparing the GRAIL-predicted cancer type of blinded samples with the known cancer identity of the samples

Results

- Of 449 total samples, 86.0% (n=386) were R/R disease and 95.5% (n=429) passed quality control
- At a prespecified detection specificity threshold of 98%, the novel GRAIL heme classifier demonstrated a high cancer detection rate of 91.8% (394/429) with a high CSO accuracy of 95.7% (377/394) overall (**Figure 1**)
 - CSO accuracy ranged from 86.8% for AML to 98.3% for MM
 - There was a high correlation of prediction to cancer type and high confidence calls up to 10^{-3} to 10^{-4} detection thresholds; an increase in cancer-positive calls was seen at lower detection specificity thresholds

The GRAIL assay reproducibly detected blood cancer

- In double baseline samples, cancer was reproducibly detected in 49 of 55 (89.1%) cases (**Figure 2** [left])
 - 4/55 (7.3%) did not have detected tumor at T0 (screening), but tumor was detected at T1 (cycle 1, day 1 predose)
 - 3 of the 4 were detected with lower specificity cutoffs, 2 of which had a correctly predicted indication
 - 2 of 55 (3.6%) were cancer-positive at T0 and cancer-negative at T1
- MVAF for each of the 55 samples is presented at both T0 and T1 (**Figure 2** [right])

LOD was between 10^{-3} to 10^{-4} across blood cancer types

- Sensitivity analyses using orthogonal MRD assays of posttreatment CLL (flow cytometry based; **Supplemental Figure 1**) and MCL (NGS based; **Figure 3**) samples suggest the current GRAIL heme classifier LOD is ~10^{-3} to 10^{-4} MVAF
- Sensitivity analyses based on serial dilutions of DLBCL (**Figure 4**) and CLL (**Supplemental Figure 2**) patient plasma samples spiked into healthy volunteer plasma samples support an LOD of 10^{-4} MVAF
- The GRAIL heme classifier yielded moderate sensitivity in post-treatment samples (**Figure 5**)
 - Methylated ctDNA was detected in the 10^{-4} to 10^{-6} range for post-treatment samples, though CIs were large
 - Results from the combined data sets (all samples) also suggest the sensitivity is within the 10^{-3} to 10^{-4} range
- At lower specificity thresholds, the classifier allowed for detection of more cancer samples (**Figure 6**) but could introduce false positives and impact sensitivity

Figure 1. GRAIL CSO Prediction Shows Selectivity in Several Hematologic Malignancy Subtypes

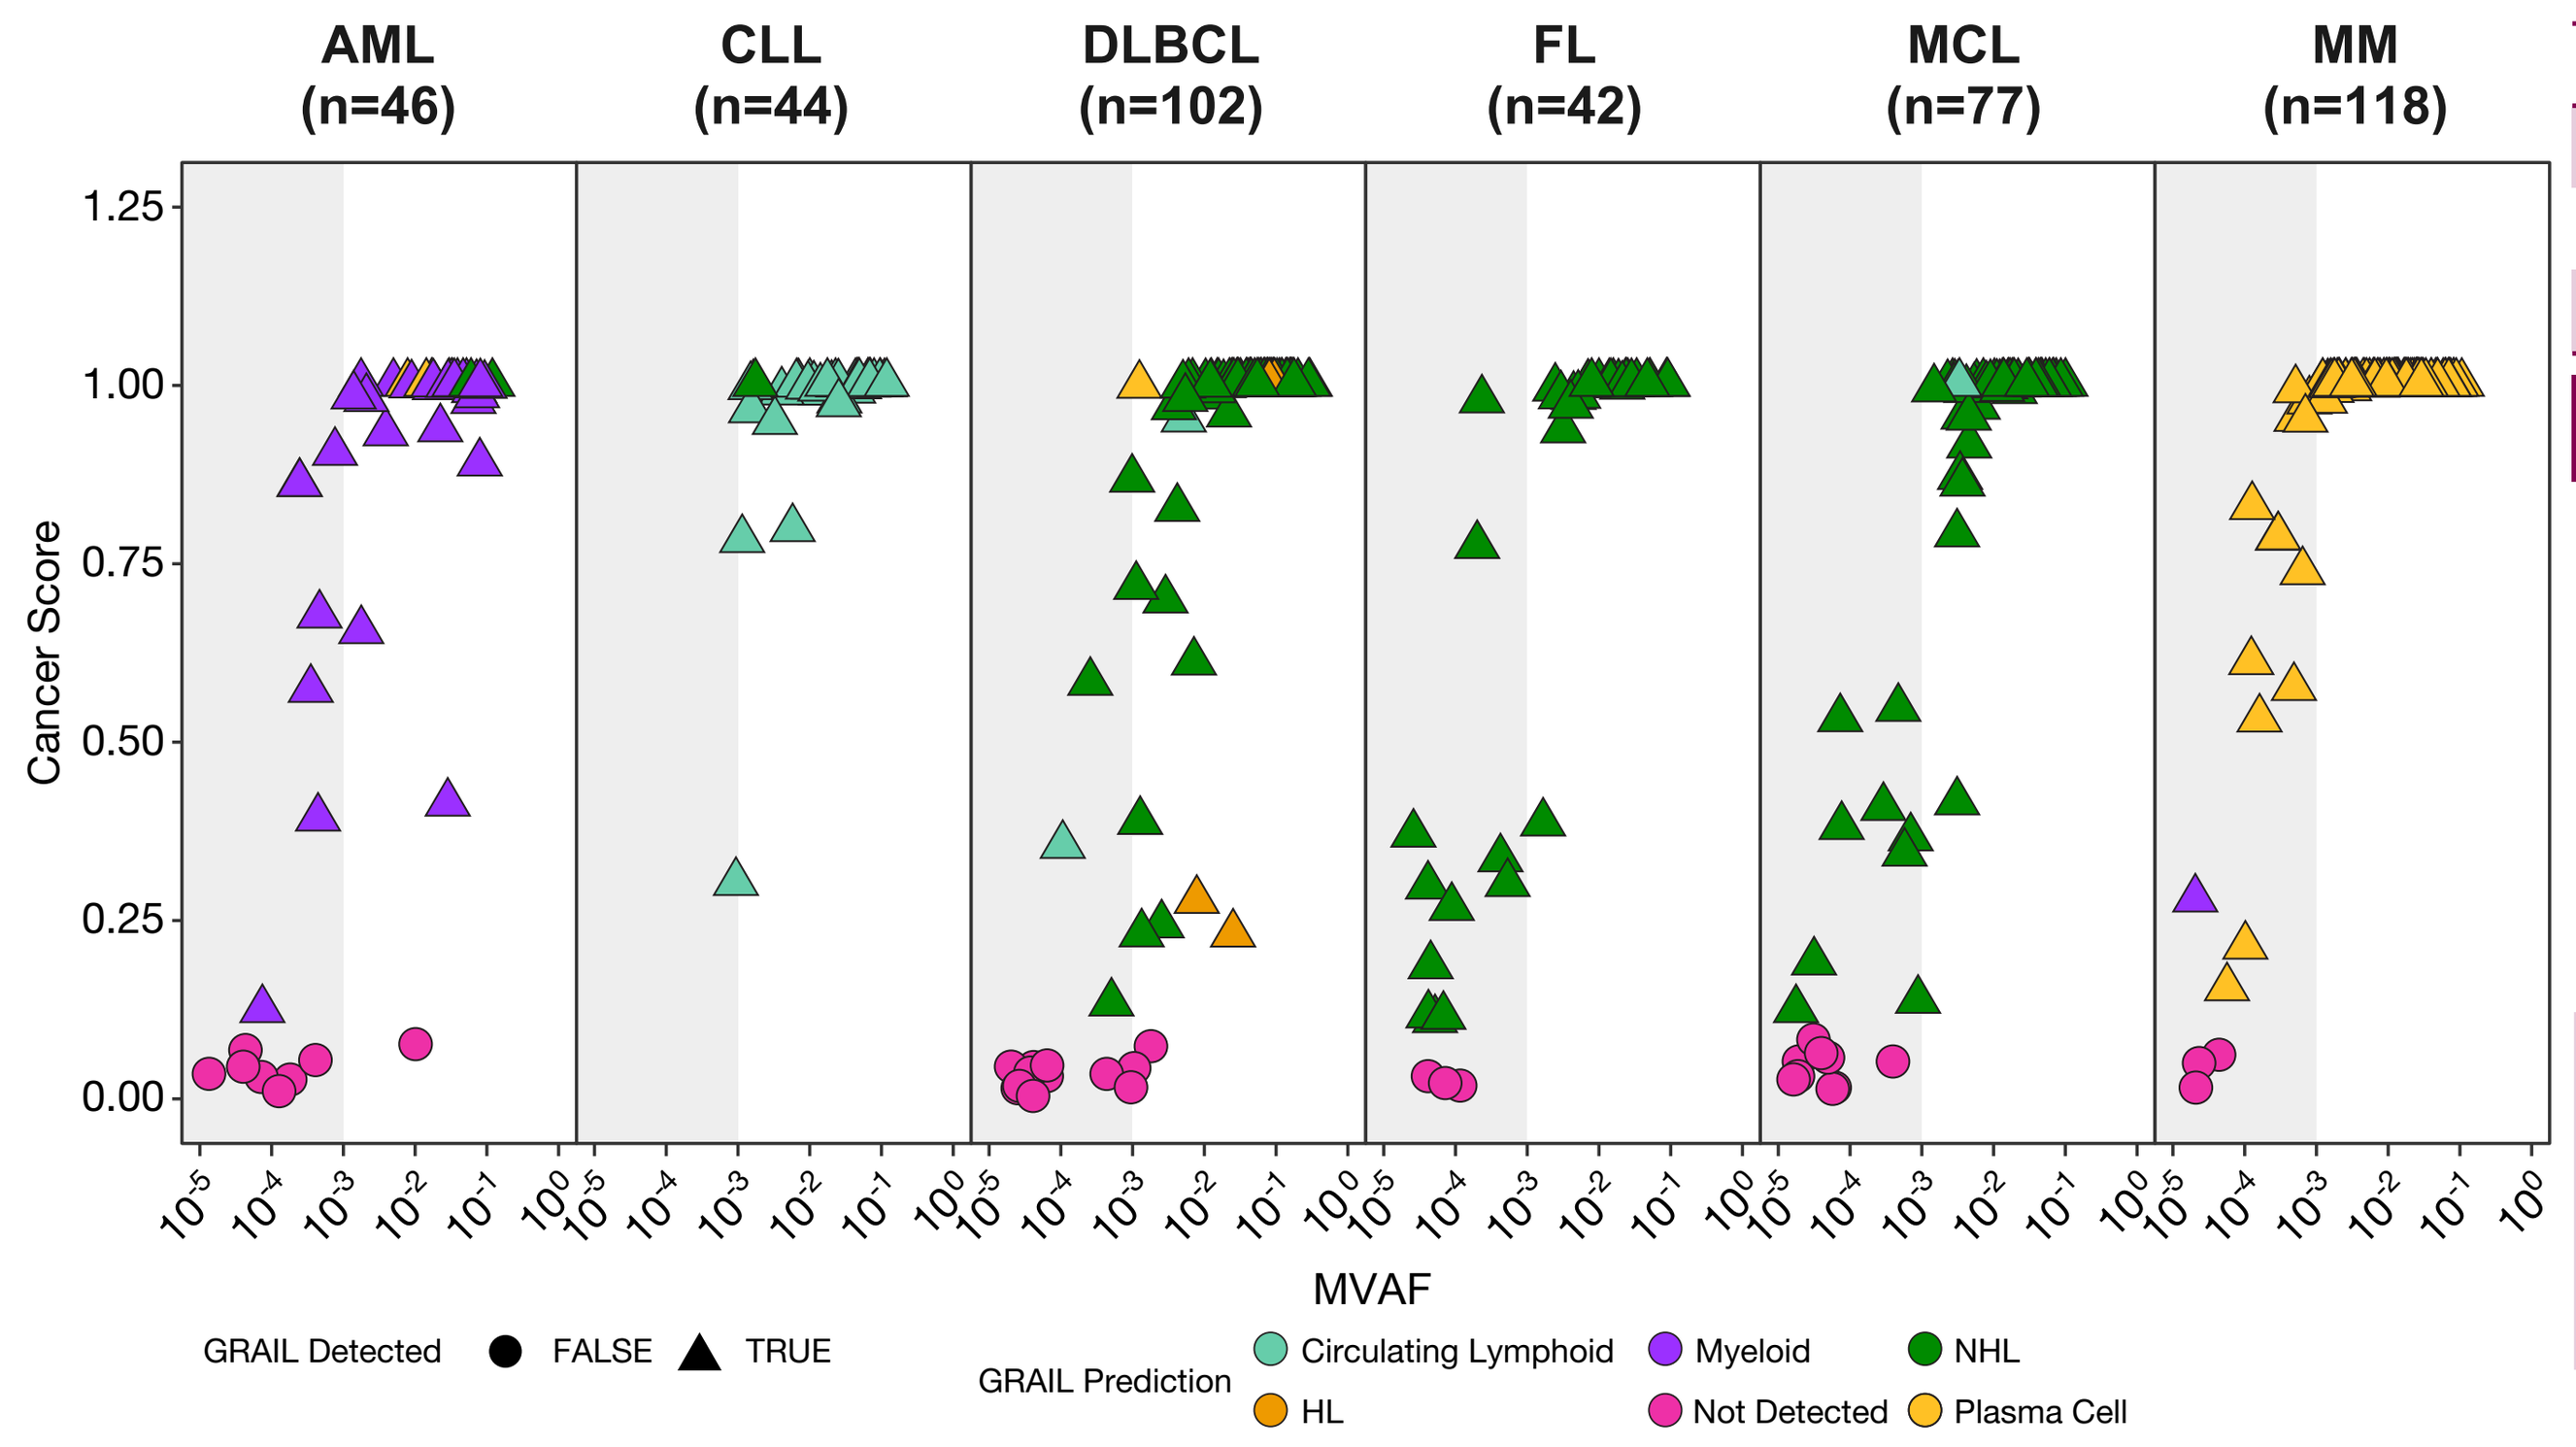
AML (n=46)
CLL (n=44)
DLBCL (n=102)
FL (n=42)
MCL (n=77)
MM (n=118)
Cancer Score
MVAF
GRAIL Detected FALSE TRUE
GRAIL Prediction Circulating Lymphoid Myeloid NHL HL Not Detected Plasma Cell

Total Samples	449	%
QC Passed	429	95.6
Cancer Detection	394	91.8
CSO	377	95.7

GRAIL Prediction

		Myeloid	Circulating Lymphoid	NHL	HL	Plasma Cell	Total	% Matched
	AML	33	0	2	0	3	38	86.8
	CLL	0	43	1	0	0	44	97.7
NHL	DLBCL	0	2	82	5	1	90	95.4
NHL	FL	0	0	39	0	0	39	
NHL	MCL	0	1	67	0	0	68	
	MM	1	0	0	1	113	115	98.3

Of the 10 frontline cases, the prediction rate was 100% for FL and 91% for MCL.

Figure 2. GRAIL Prediction (Left) and MVAF (Right) in Double Baseline Samples

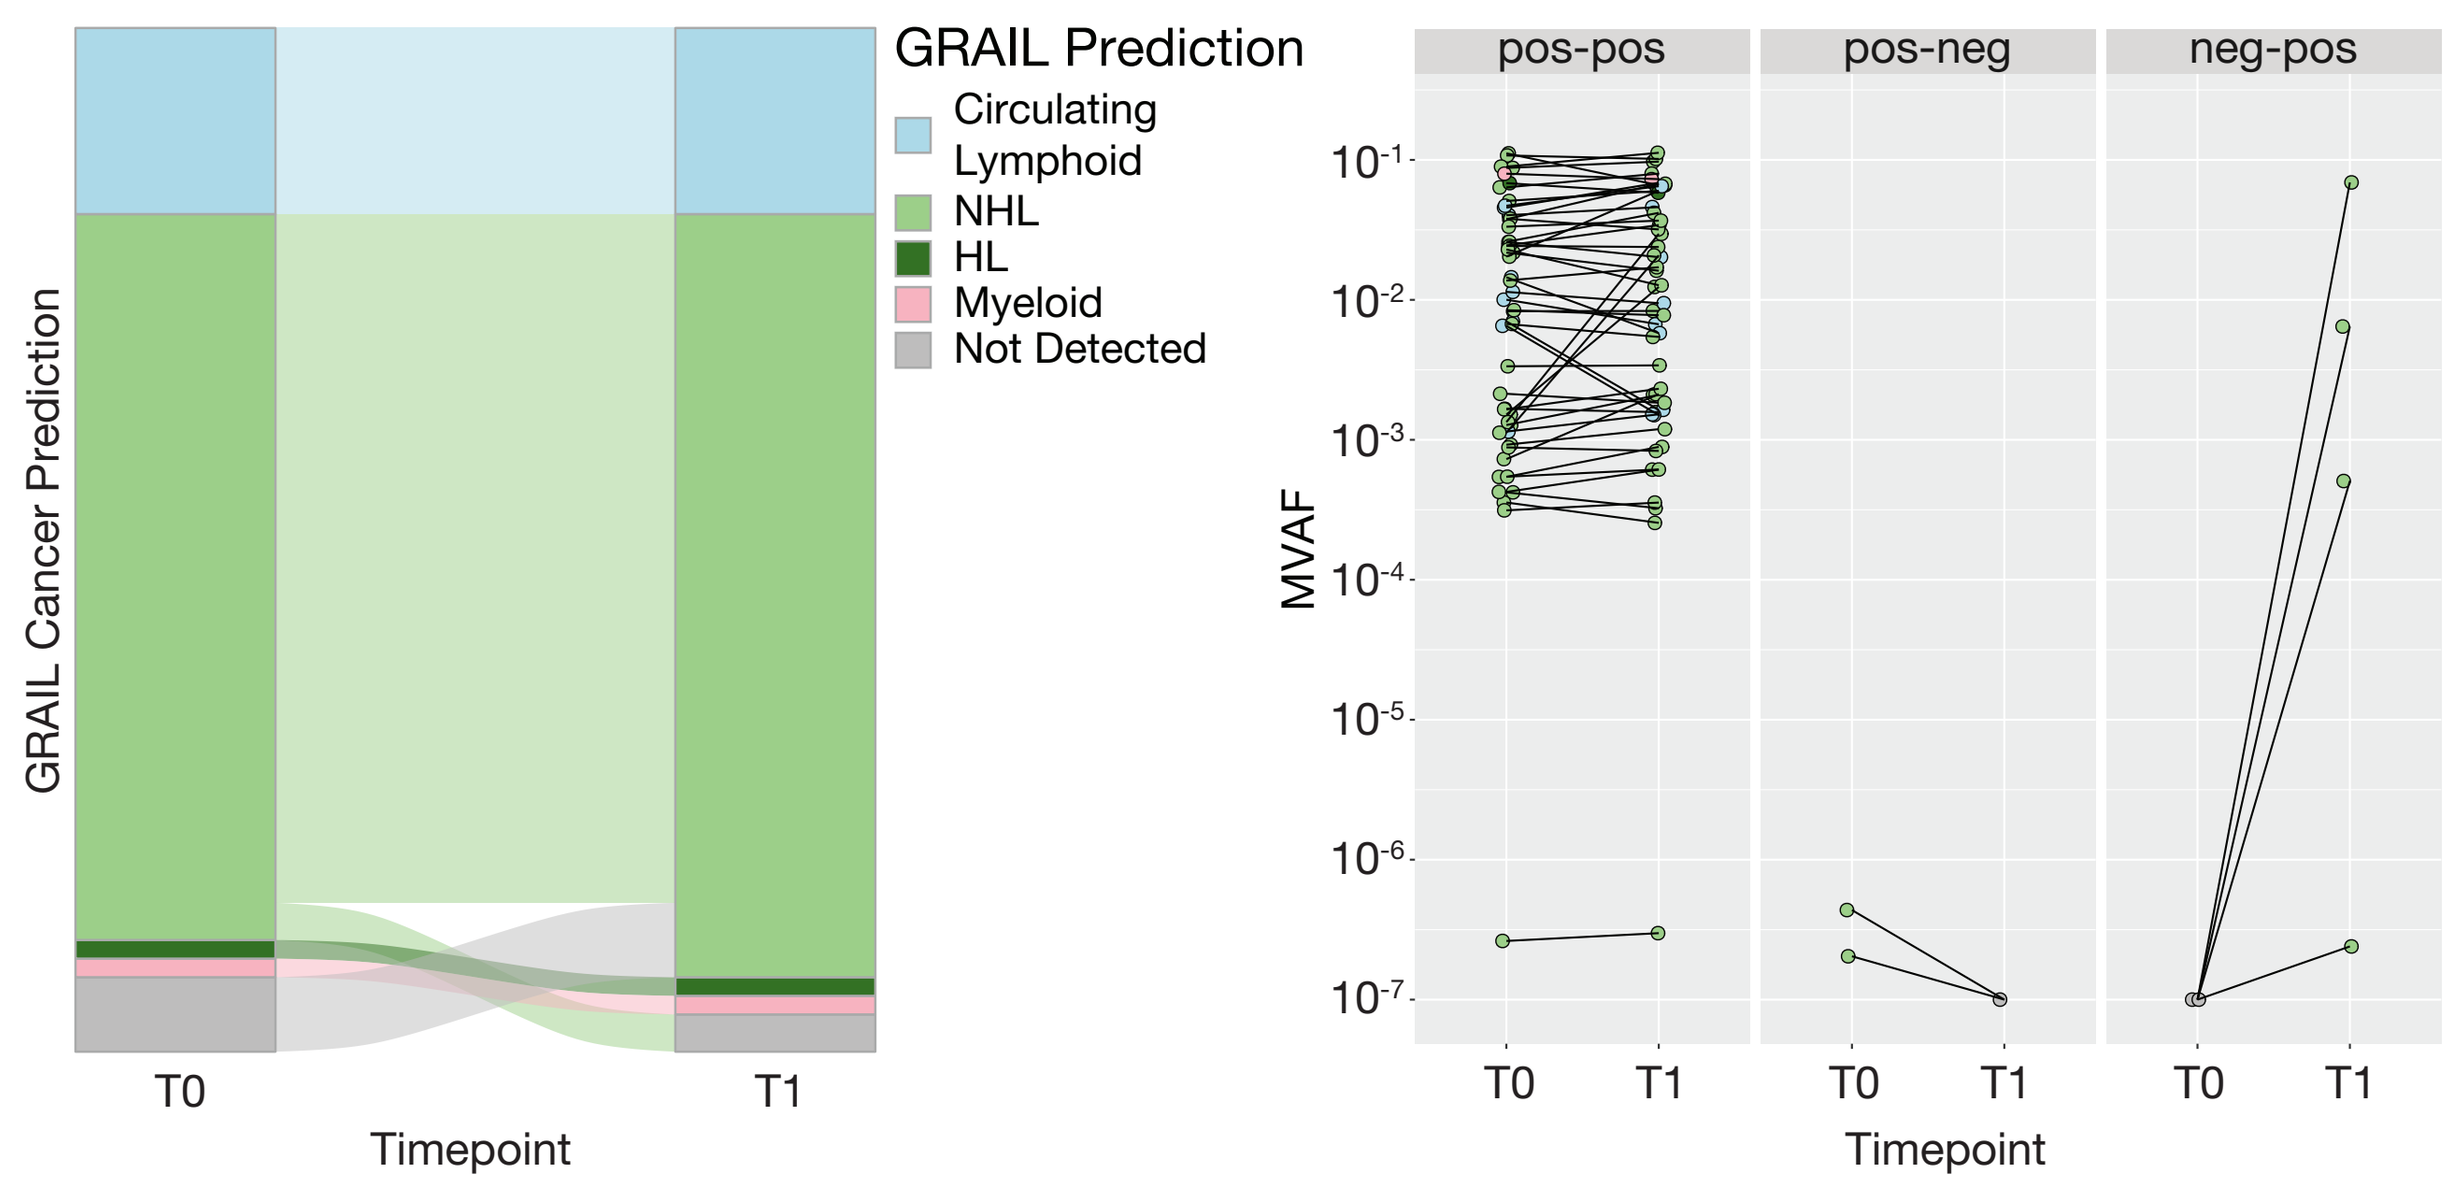
GRAIL Prediction
Circulating Lymphoid
NHL
HL
Myeloid
Not Detected
GRAIL Cancer Prediction
T0
T1
Timepoint
pos-pos
pos-neg
neg-pos
MVAF

GRAIL prediction of double baseline samples at the default specificity threshold of 98%.

Figure 3. Orthogonal GRAIL Correlation in MCL ClonoSEQ Samples

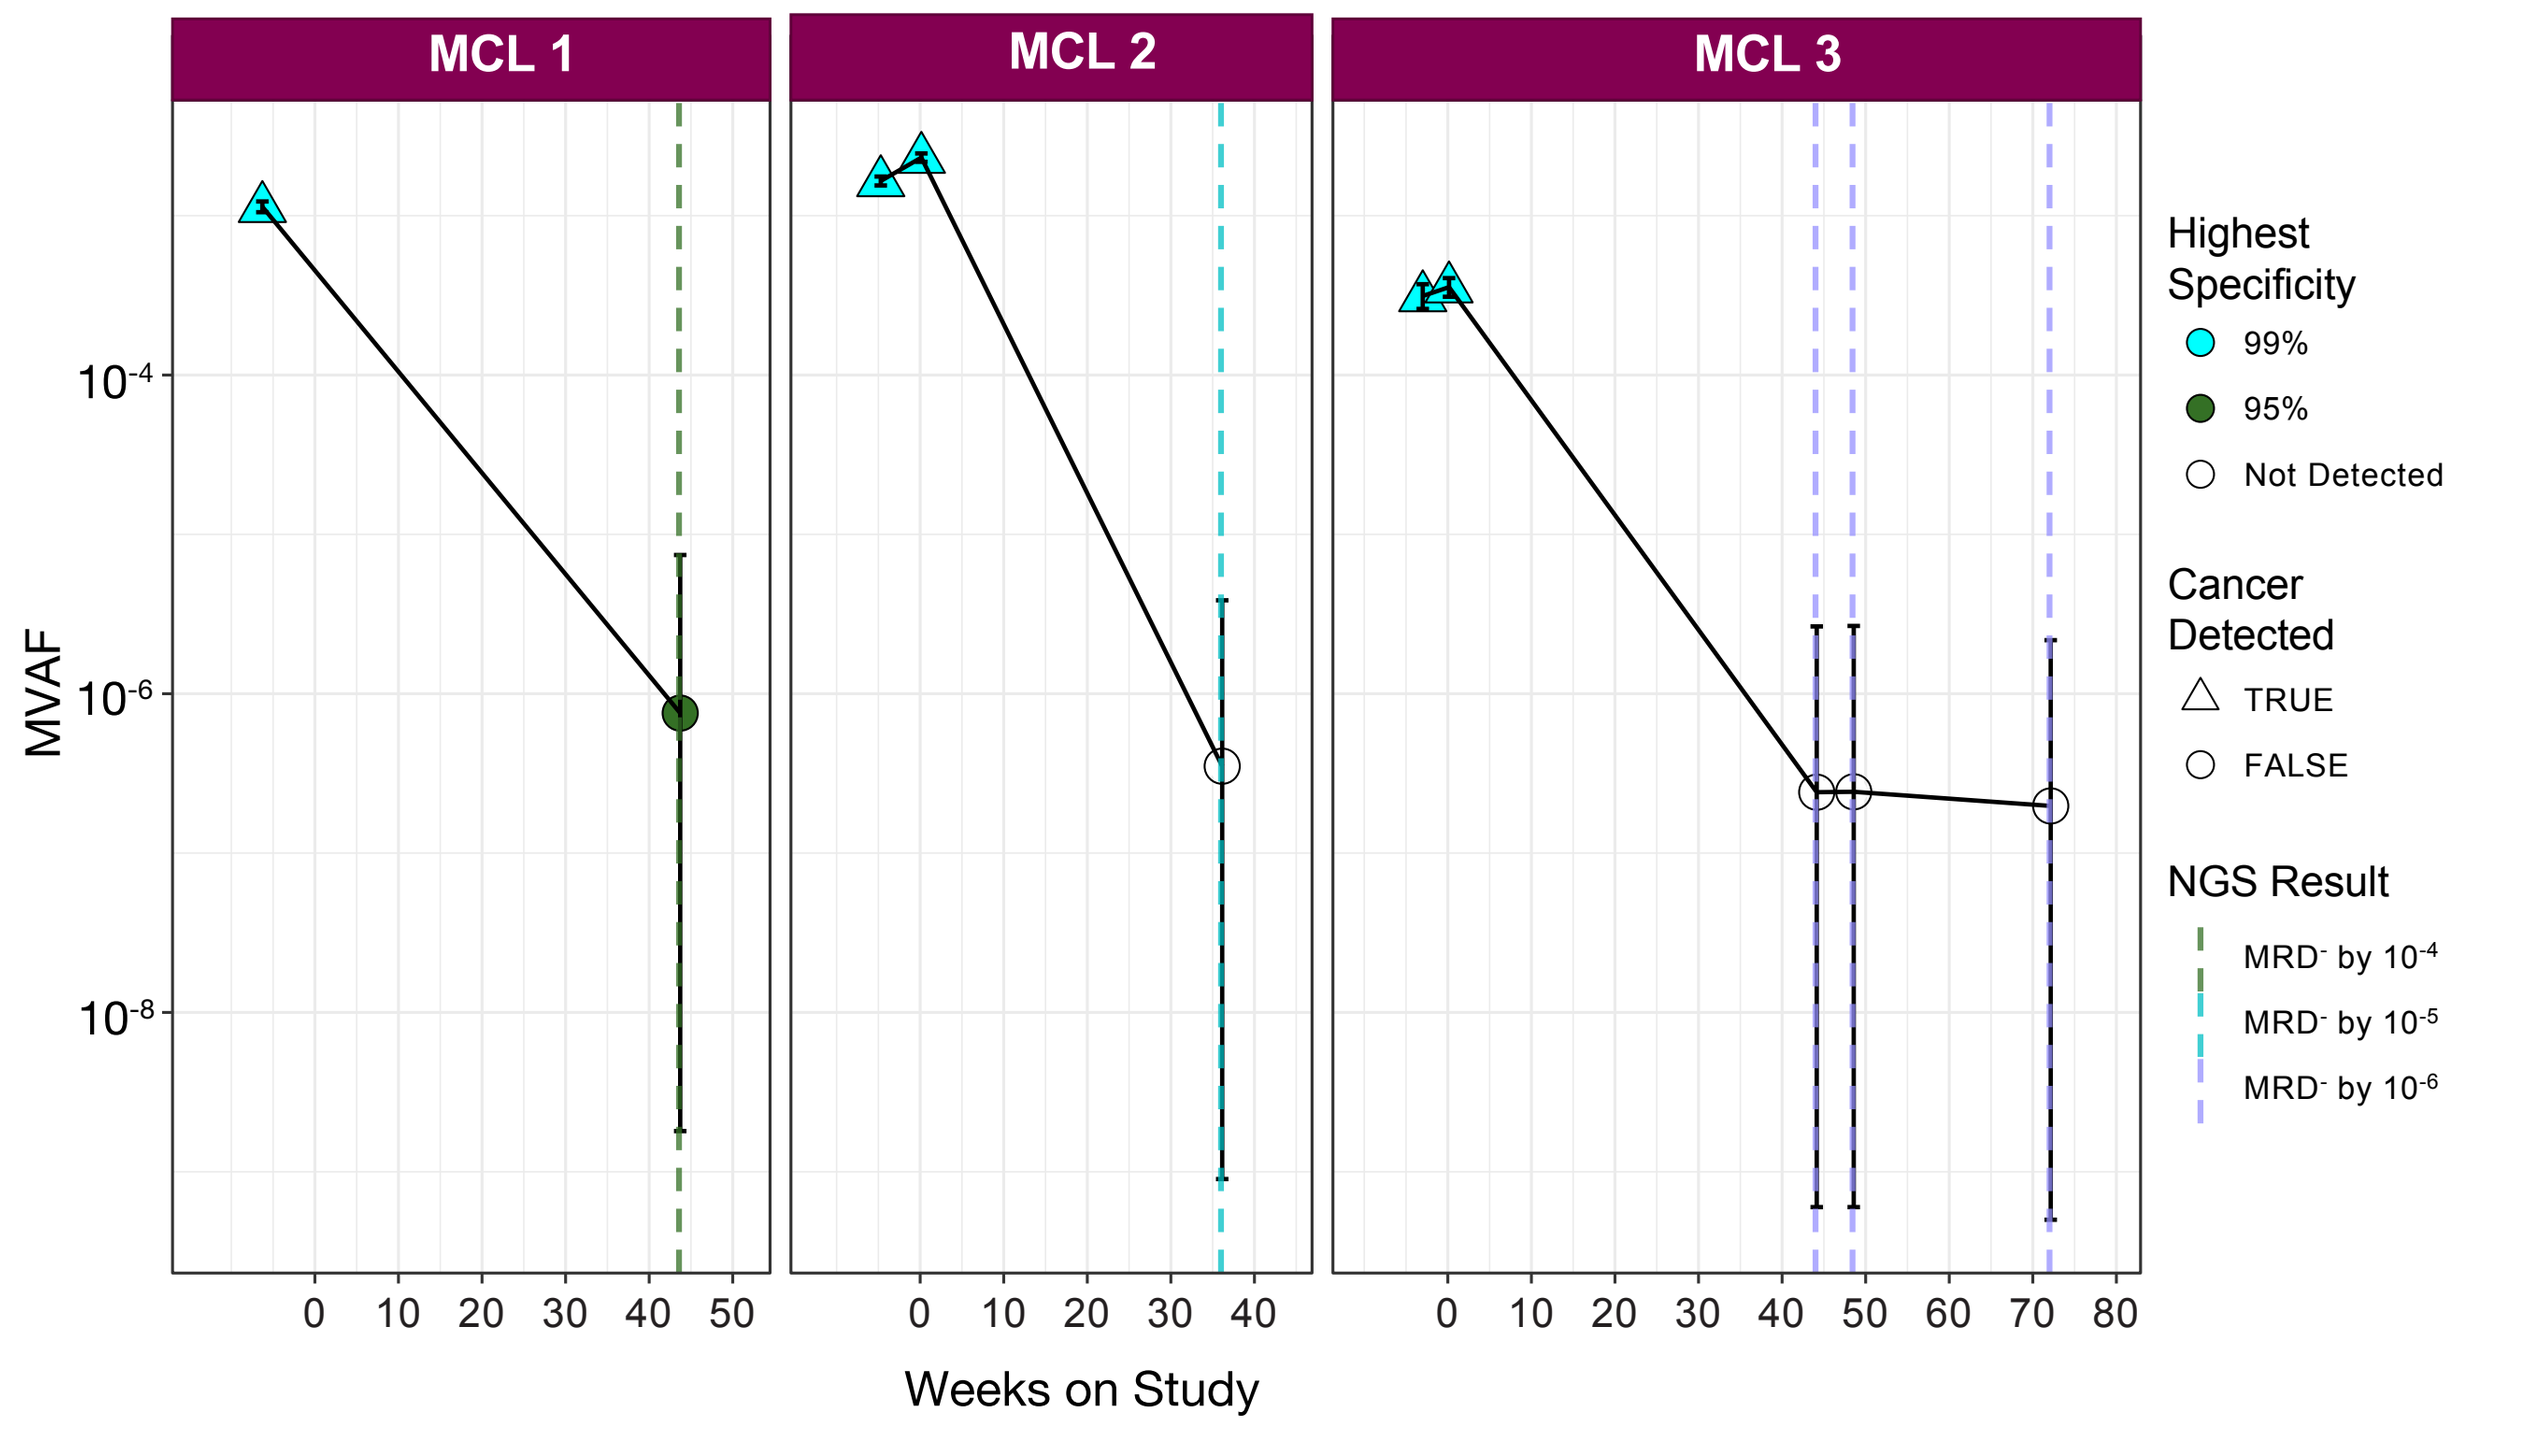
MCL 1
MCL 2
MCL 3
MVAF
Weeks on Study
Highest Specificity
99%
95%
Not Detected
Cancer Detected
TRUE
FALSE
NGS Result

Error bars represent 95% CI. Orthogonal data were based on cell-based assays.

Figure 4. GRAIL Classifier on Serial Dilutions of DLBCL Patient Plasma Samples

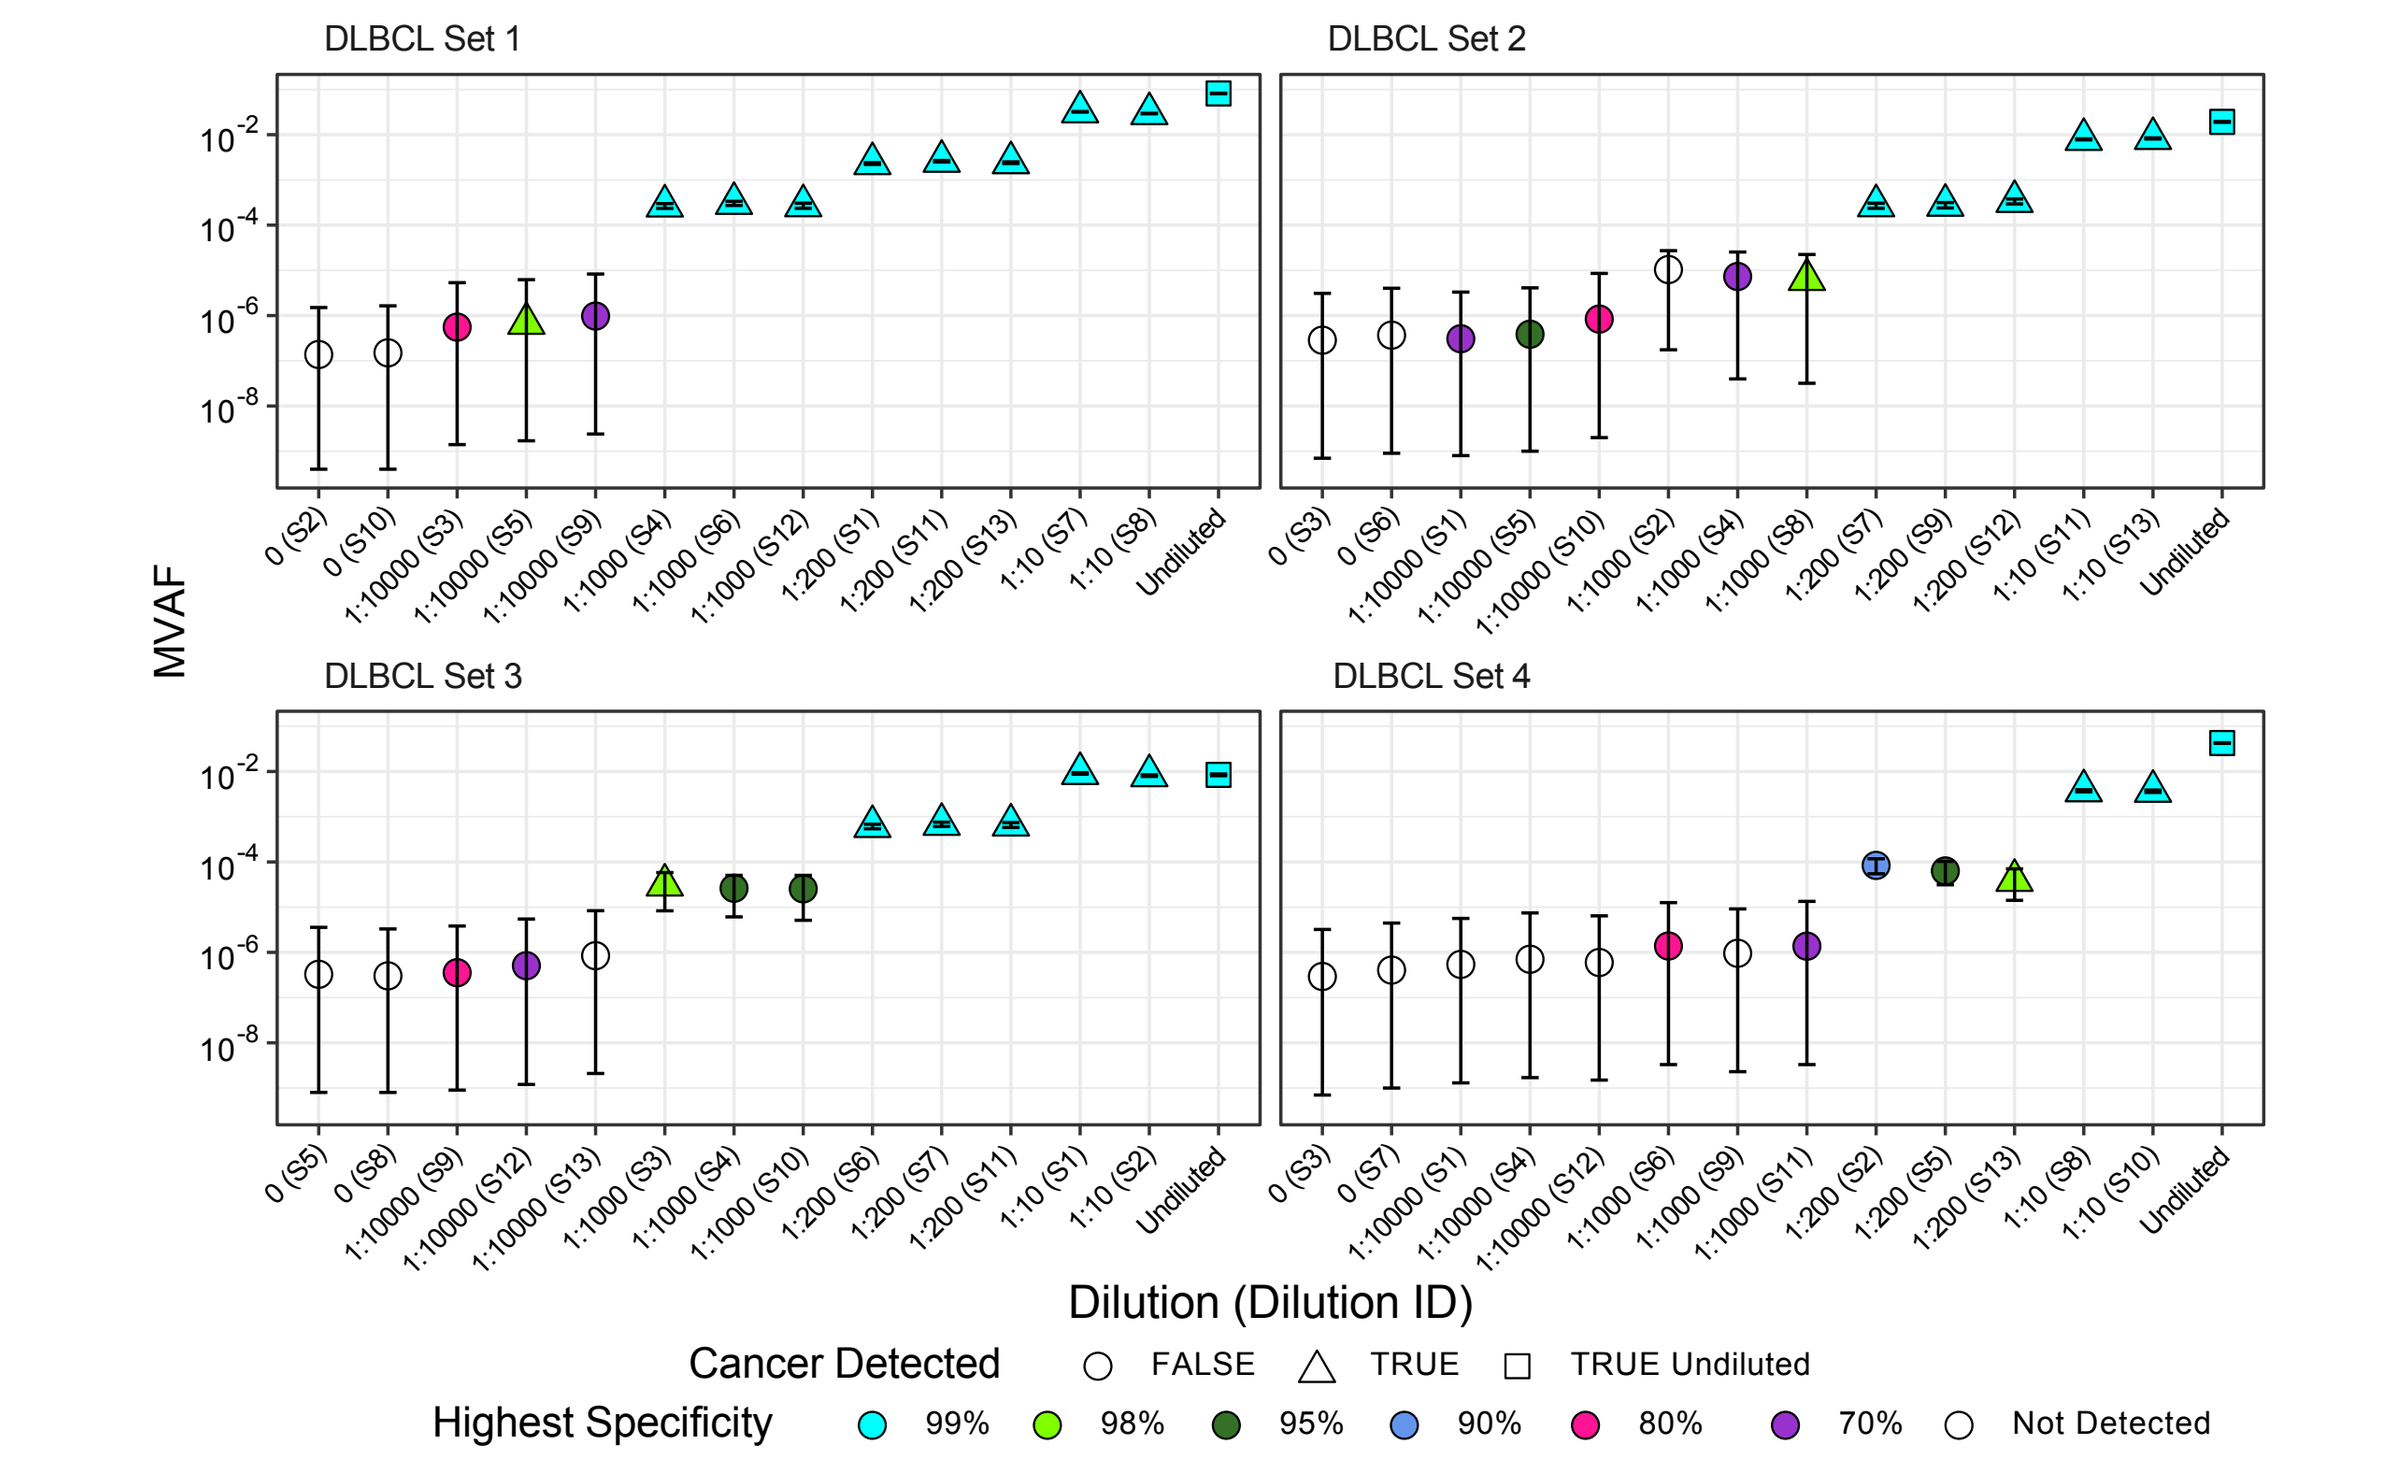
DLBCL Set 1
DLBCL Set 2
DLBCL Set 3
DLBCL Set 4
MVAF
Dilution (Dilution ID)
Cancer Detected FALSE TRUE TRUE Undiluted
Highest Specificity 99% 98% 95% 90% 80% 70% Not Detected

Error bars represent 95% CI.

Figure 5. GRAIL Algorithm in All* Samples Tested

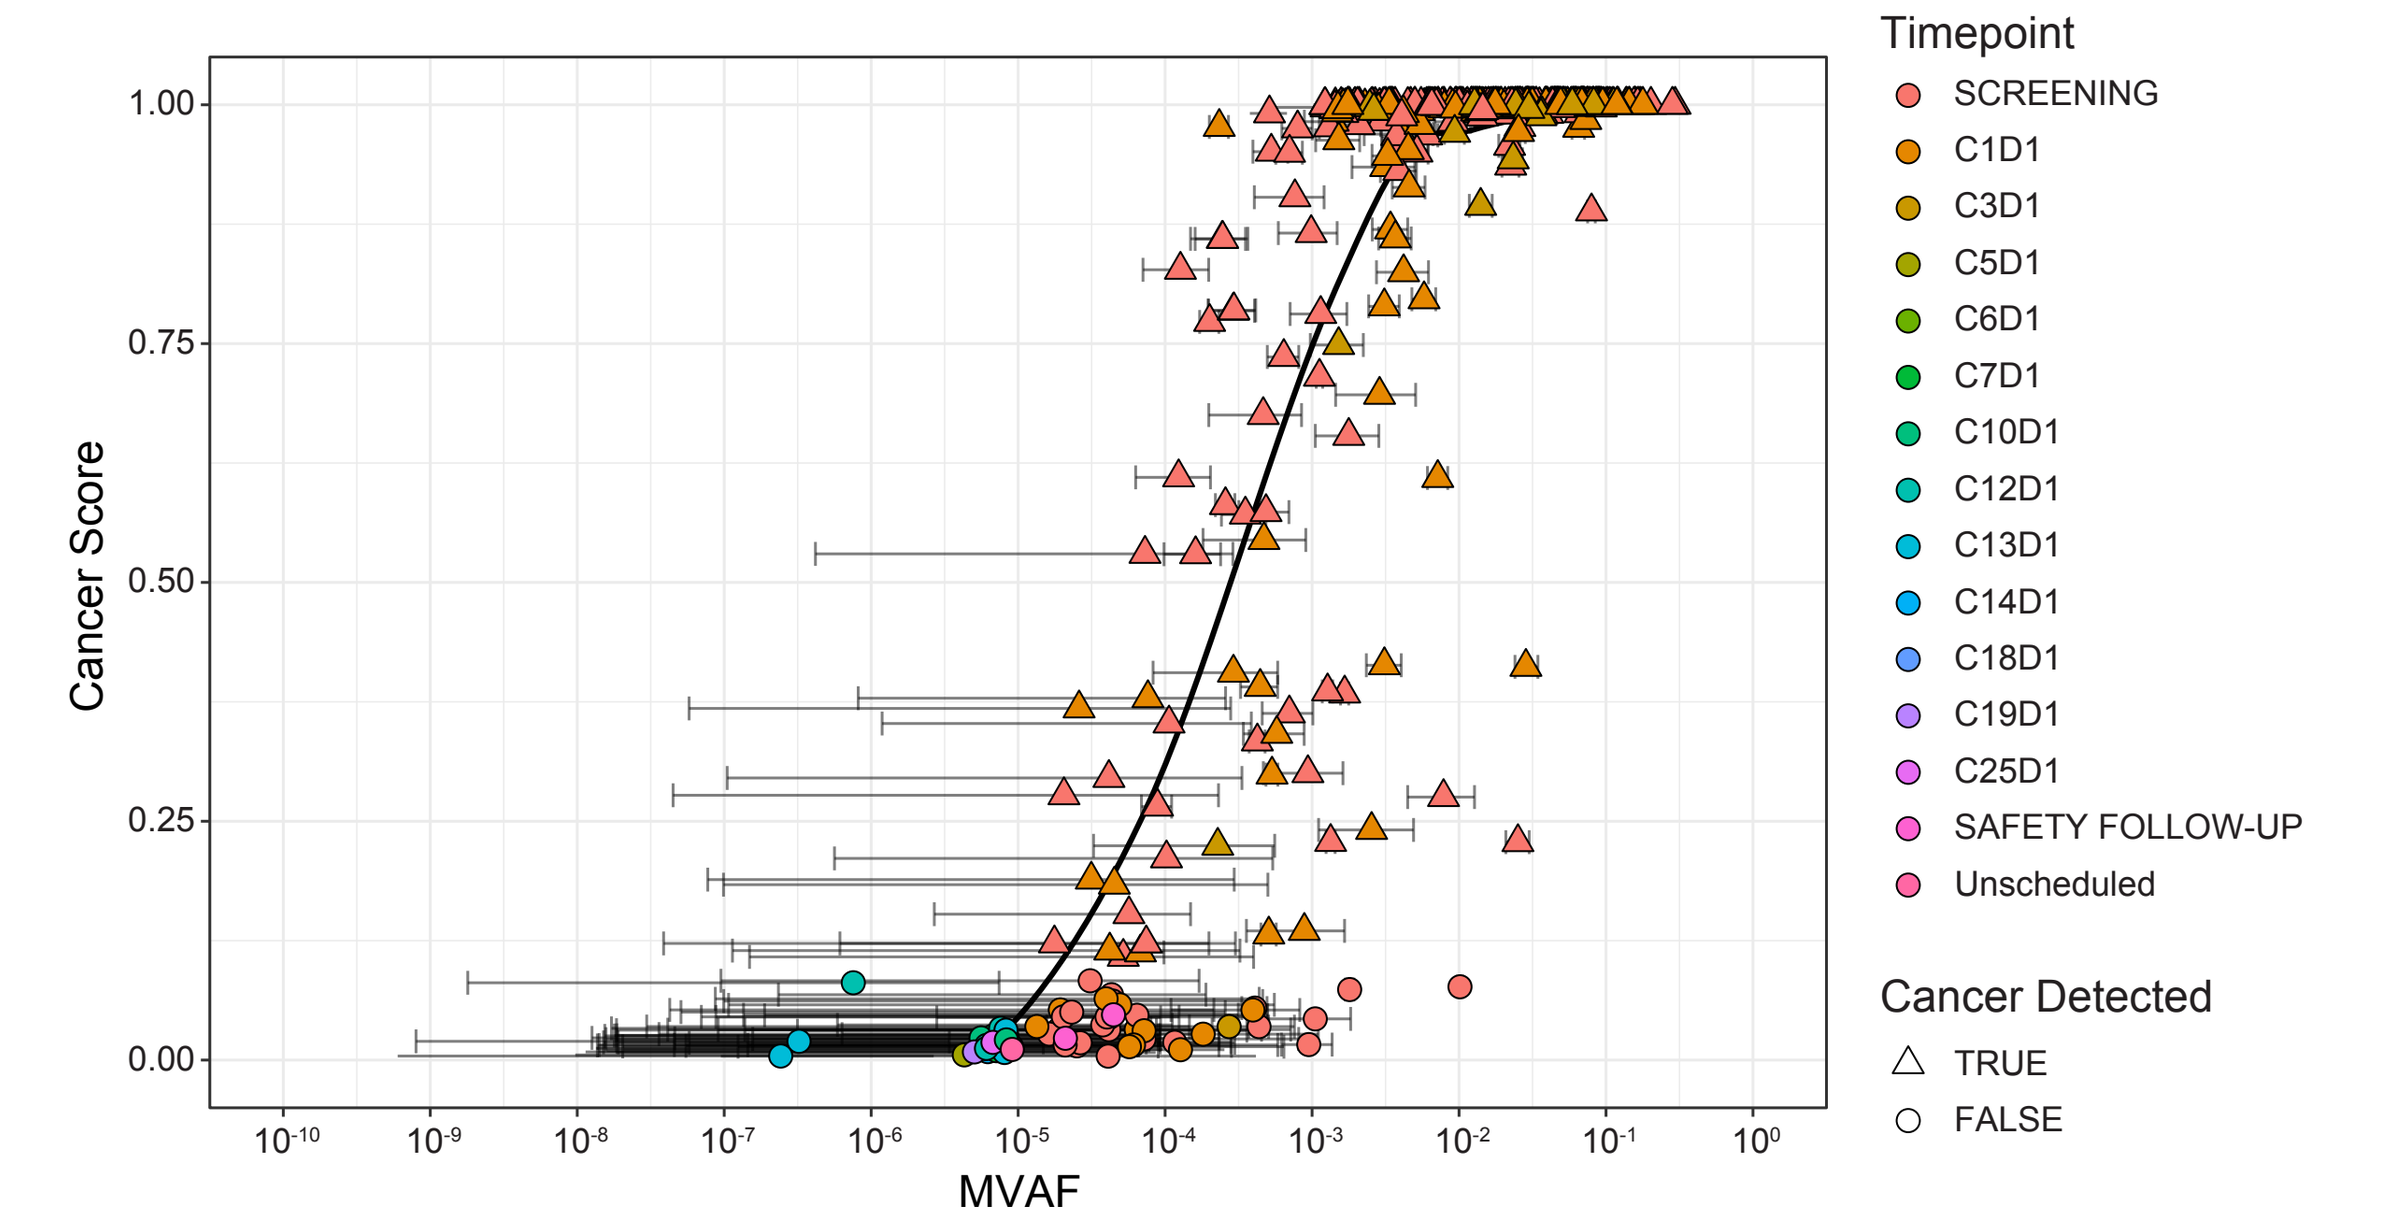
Cancer Score
MVAF
Timepoint
SCREENING
C1D1
C3D1
C5D1
C6D1
C7D1
C10D1
C12D1
C13D1
C14D1
C18D1
C19D1
C25D1
SAFETY FOLLOW-UP
Unscheduled
Cancer Detected
TRUE
FALSE

*Includes baseline and post-treatment samples. Error bars represent 95% CI.

Figure 6. Cancer Detection Rate Concordance Across Different Classifier Specificities

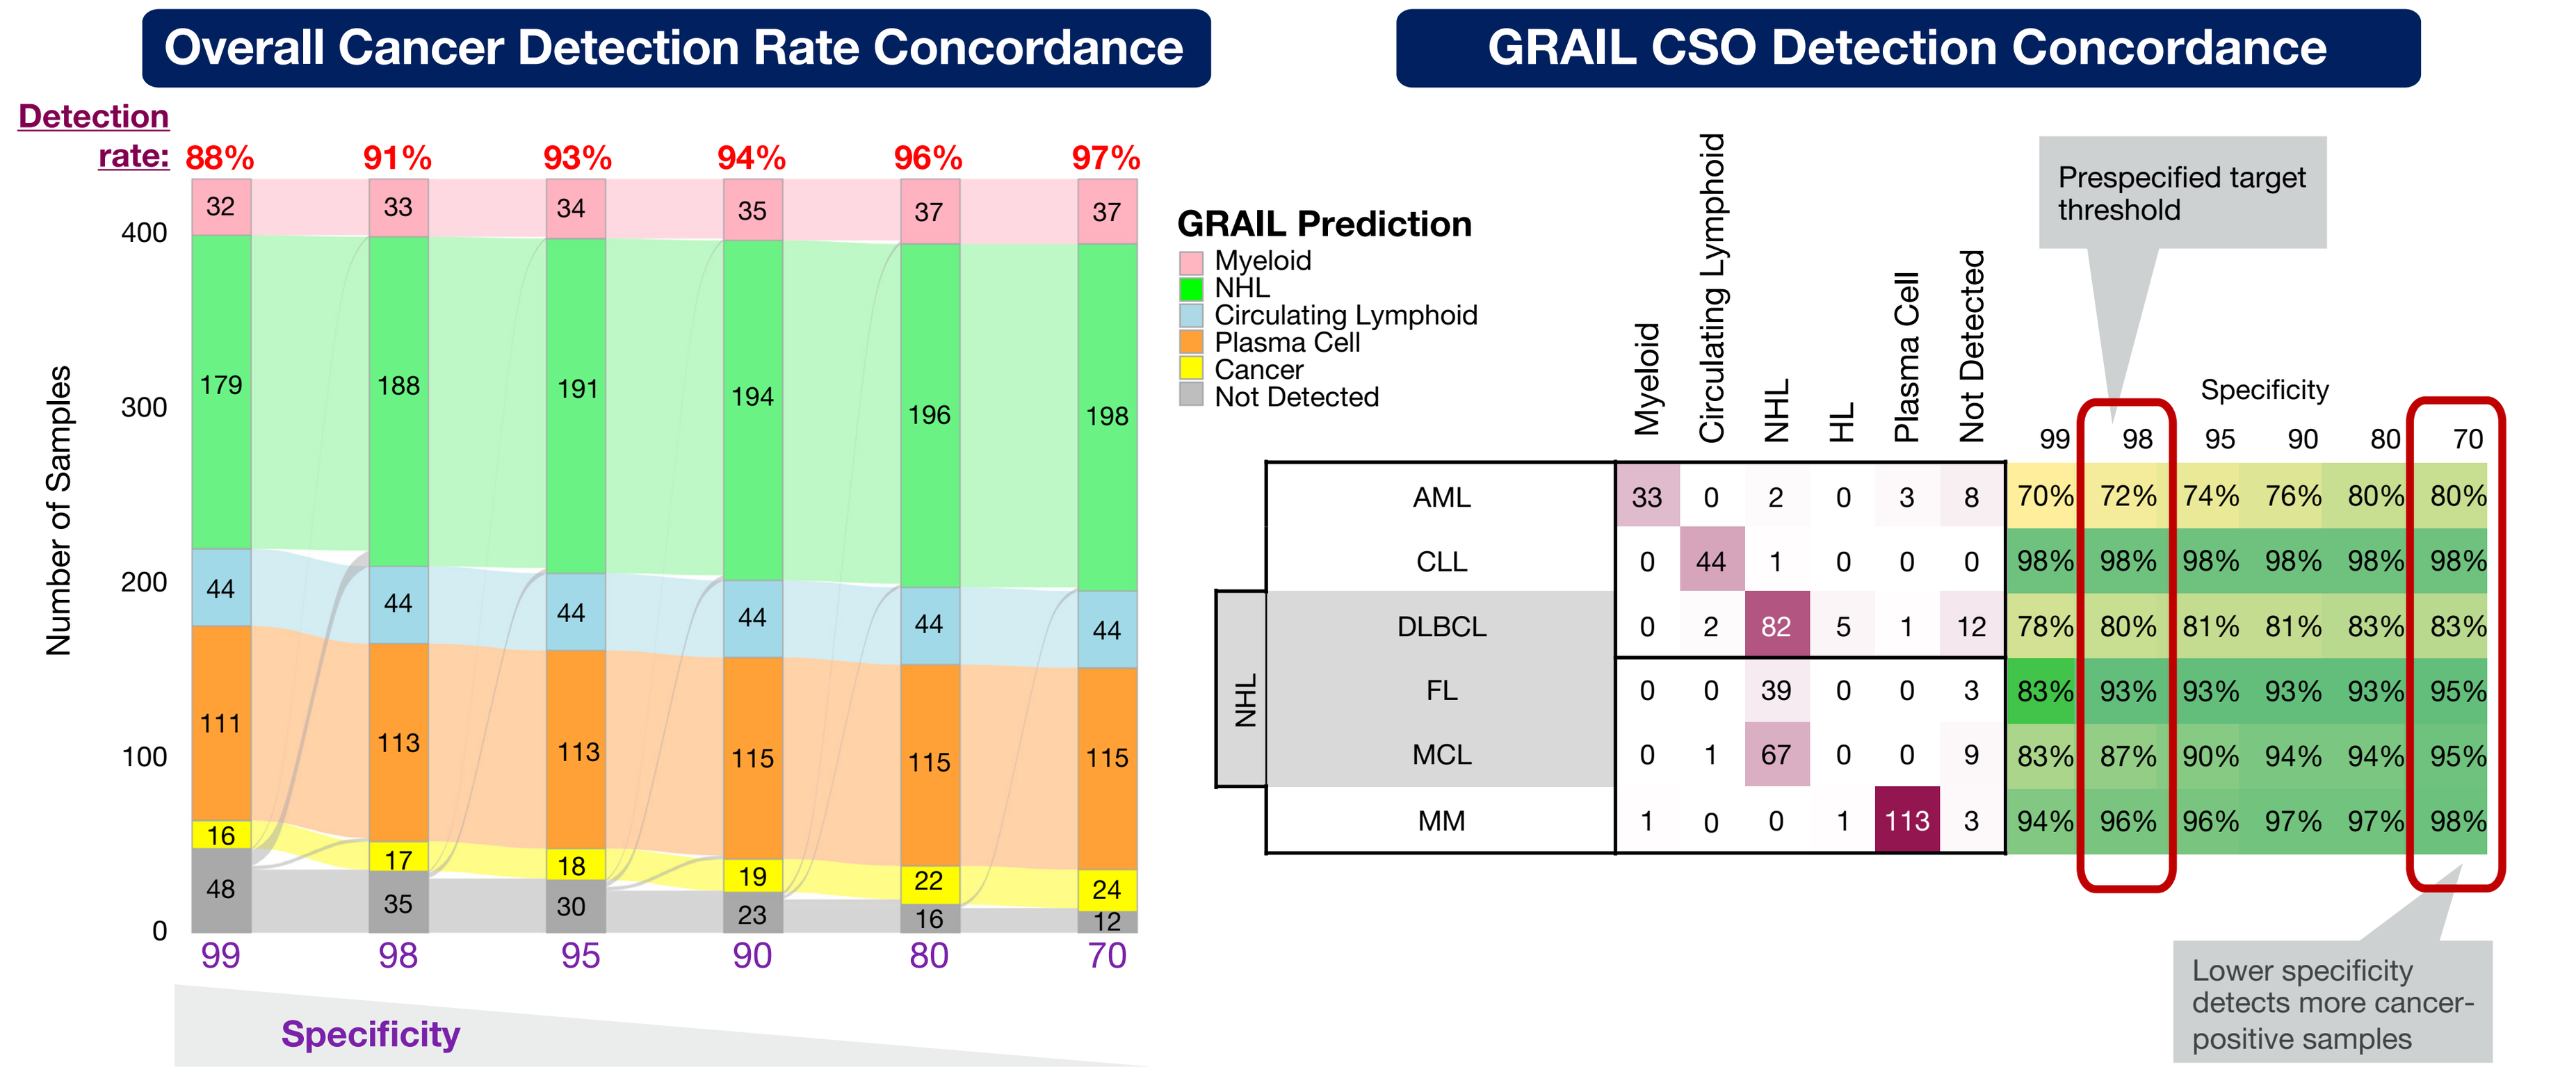
Overall Cancer Detection Rate Concordance
GRAIL CSO Detection Concordance
Detection rate: 88% 91% 93% 94% 96% 97%
Number of Samples
Specificity
GRAIL Prediction
Myeloid
NHL
Circulating Lymphoid
Plasma Cell
Cancer
Not Detected
Prespecified target threshold
Lower specificity detects more cancer-positive samples

Conclusions

- Findings suggest that the GRAIL heme classifier can identify cancer signal from patients with R/R disease across multiple hematologic malignancies
 - 91.8% cancer detection rate, 95.7% CSO rate
 - Of the detected samples, CSO was highly accurate: 97.7% in CLL, 98.3% in MM, and 95.4% in non-Hodgkin lymphoma (DLBCL, FL, and MCL)
- Orthogonal MRD assays indicated a GRAIL classifier LOD of ~10^{-3} to 10^{-4} MVAF
- The GRAIL heme classifier demonstrated high biologic reproducibility
- The GRAIL heme classifier is currently in development; its performance characteristics have not yet been determined by GRAIL. The heme assay has not been cleared or approved by the US Food and Drug Administration
- Overall, the results support further development of a blood-based, tumor-agnostic, methylated ctDNA MRD assay with potential utility in several hematologic indications

References

1. Riva G, et al. *Cancers*. 2021;13:4582. **2.** Short NJ, et al. *Leukemia*. 2022;2817-26. **3.** Dix C, et al. *J Clin Med*. 2020:9;1714. **4.** Jamshidi A, et al. *Cancer Cell*. 2022;40:1537-49. **5.** Bozic T, et al. *Leukemia*. 2022;36:80-9. **6.** Kalinkova L, et al. *Int J Mol Sci*. 2023;24:633. **7.** Melton C, et al. *J Clin Oncol*. 2020;38(suppl 15): Abstr 3052. 10.1200/JCO.2020.38.15_suppl.3052.

Abbreviations

AML, acute myeloid leukemia; C, cycle; CI, confidence interval; CLL, chronic lymphocytic leukemia; CSO, cancer signal origin; ctDNA, circulating tumor DNA; D, day; DLBCL, diffuse large B-cell lymphoma; FL, follicular lymphoma; HL, Hodgkin lymphoma; LOD, limit of detection; MCL, mantle cell lymphoma; MM, multiple myeloma; MRD, minimal or measurable residual disease; MVAF, methyl variant allele fraction; NGS, next-generation sequencing; NHL, non-Hodgkin lymphoma; QC, quality control; R/R, relapsed/refractory

Acknowledgments

We thank all the patients, their families, and the investigators and their study teams who participated in the clinical trials from which plasma samples were obtained. This study was funded by AstraZeneca. Medical writing support was provided by Maria Ali, PhD, of OPEN Health, and funded by AstraZeneca. The authors directed development of the presentation and are fully responsible for all content and editorial decisions.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster Supplementary material

Corresponding author email address: veerendra.munugalavadla@astrazeneca.com

Presented at the American Association for Cancer Research (AACR) Annual Meeting; April 14–19, 2023

Utility of ctDNA-based Targeted Methylation MRD Assay for Hematologic Malignancies

Supplemental Information

Plain Language Summary

Why was this study done?

Recently, innovative approaches for measuring residual cancer cells remaining in the blood after cancer treatment have emerged. Because changes in DNA methylation patterns occur in cancer, DNA methylation could be used to identify residual cancer cells. Here, we evaluate a methylated DNA detection classifier developed by GRAIL, LLC, for its ability to identify various blood cancers accurately.

How were the data collected?

Blood samples were collected from patients with various types of blood cancers and assessed using the GRAIL heme classifier.

What were the results?

The GRAIL heme classifier correctly identified the type of blood cancer present in 87.9% of the 429 blood samples from various blood cancers tested.

Why do the results matter to patients and physicians?

A blood-based detection method would reduce the need for tumor biopsies and allow for minimally invasive, repeatable evaluations that are less burdensome to patients. Additionally, with around 160 different subtypes of blood cancers, a single classifier that can be used across all indications could simplify testing and potentially reduce costs.

Supplemental Figure 1. Orthogonal GRAIL Correlation in CLL Flow Cytometry Samples

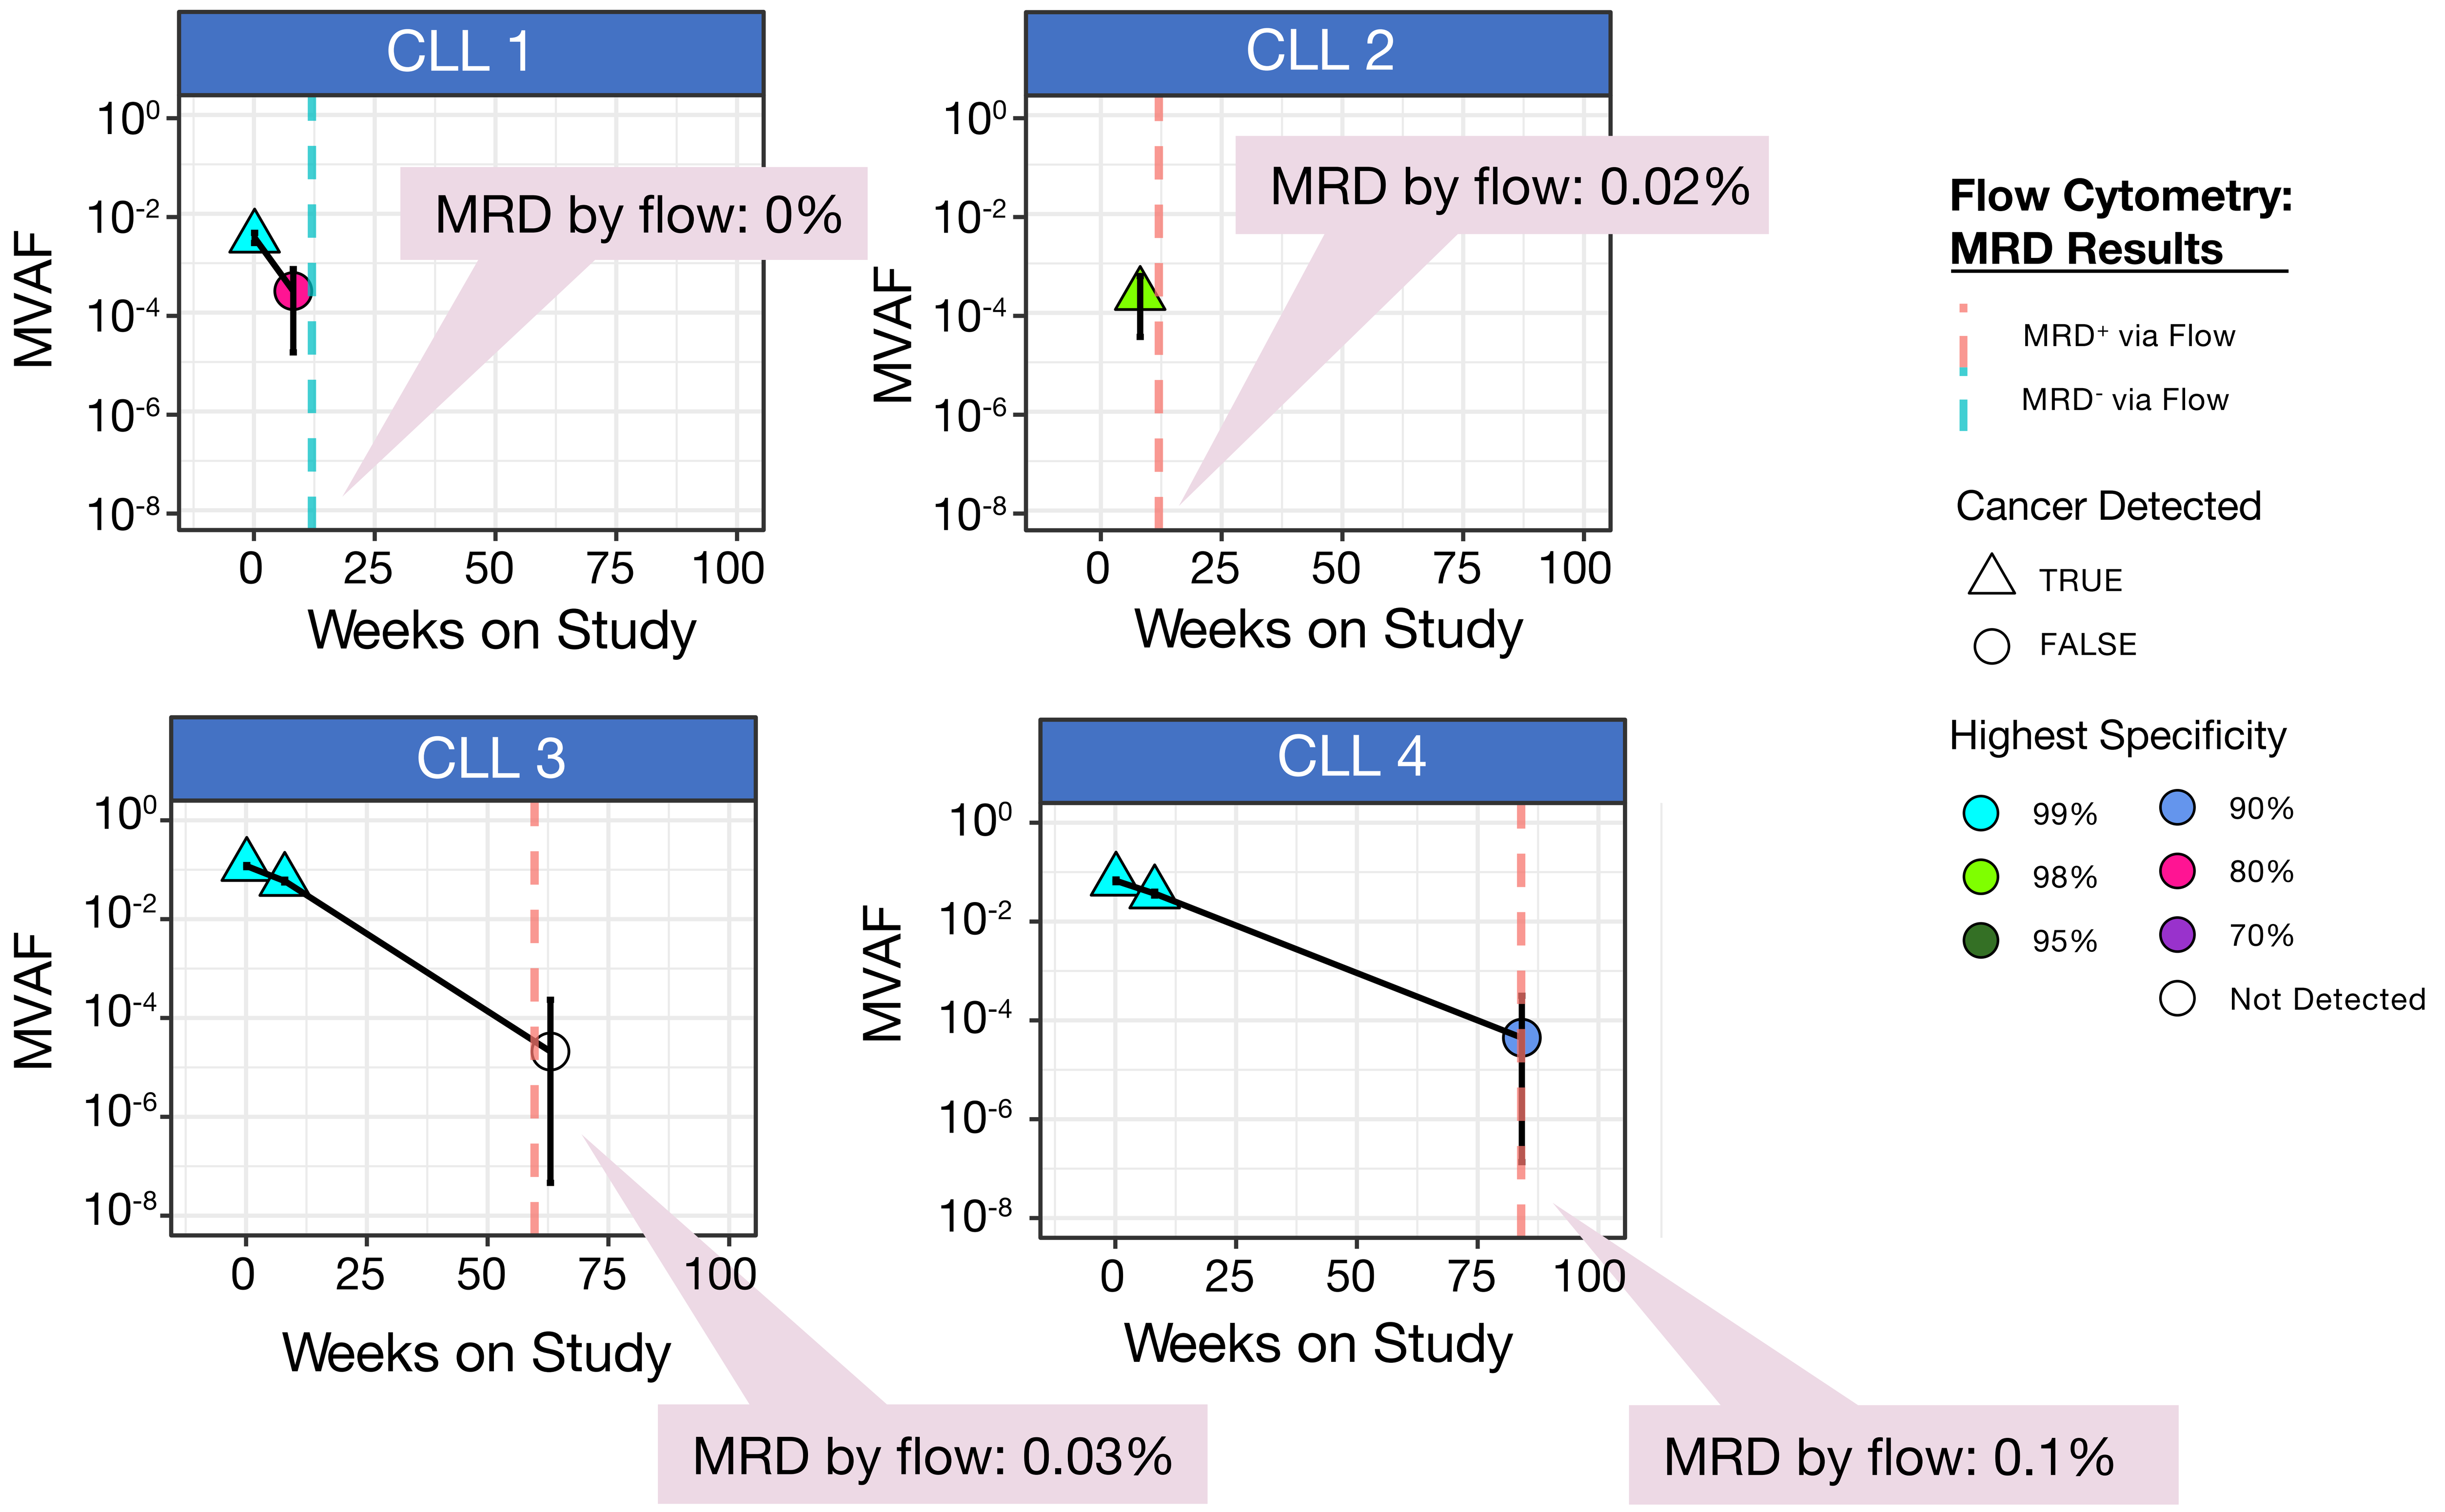

CLL 1
CLL 2
CLL 3
CLL 4
MVAF
Weeks on Study
MRD by flow: 0%
MRD by flow: 0.02%
MRD by flow: 0.03%
MRD by flow: 0.1%
Flow Cytometry: MRD Results
MRD+ via Flow
MRD- via Flow
Cancer Detected
TRUE
FALSE
Highest Specificity
99%
98%
95%
90%
80%
70%
Not Detected

Error bars represent 95% CI. Orthogonal data were based on cell-based assays.

Supplemental Figure 2. GRAIL Classifier on Serial Dilutions of CLL Patient Plasma Samples

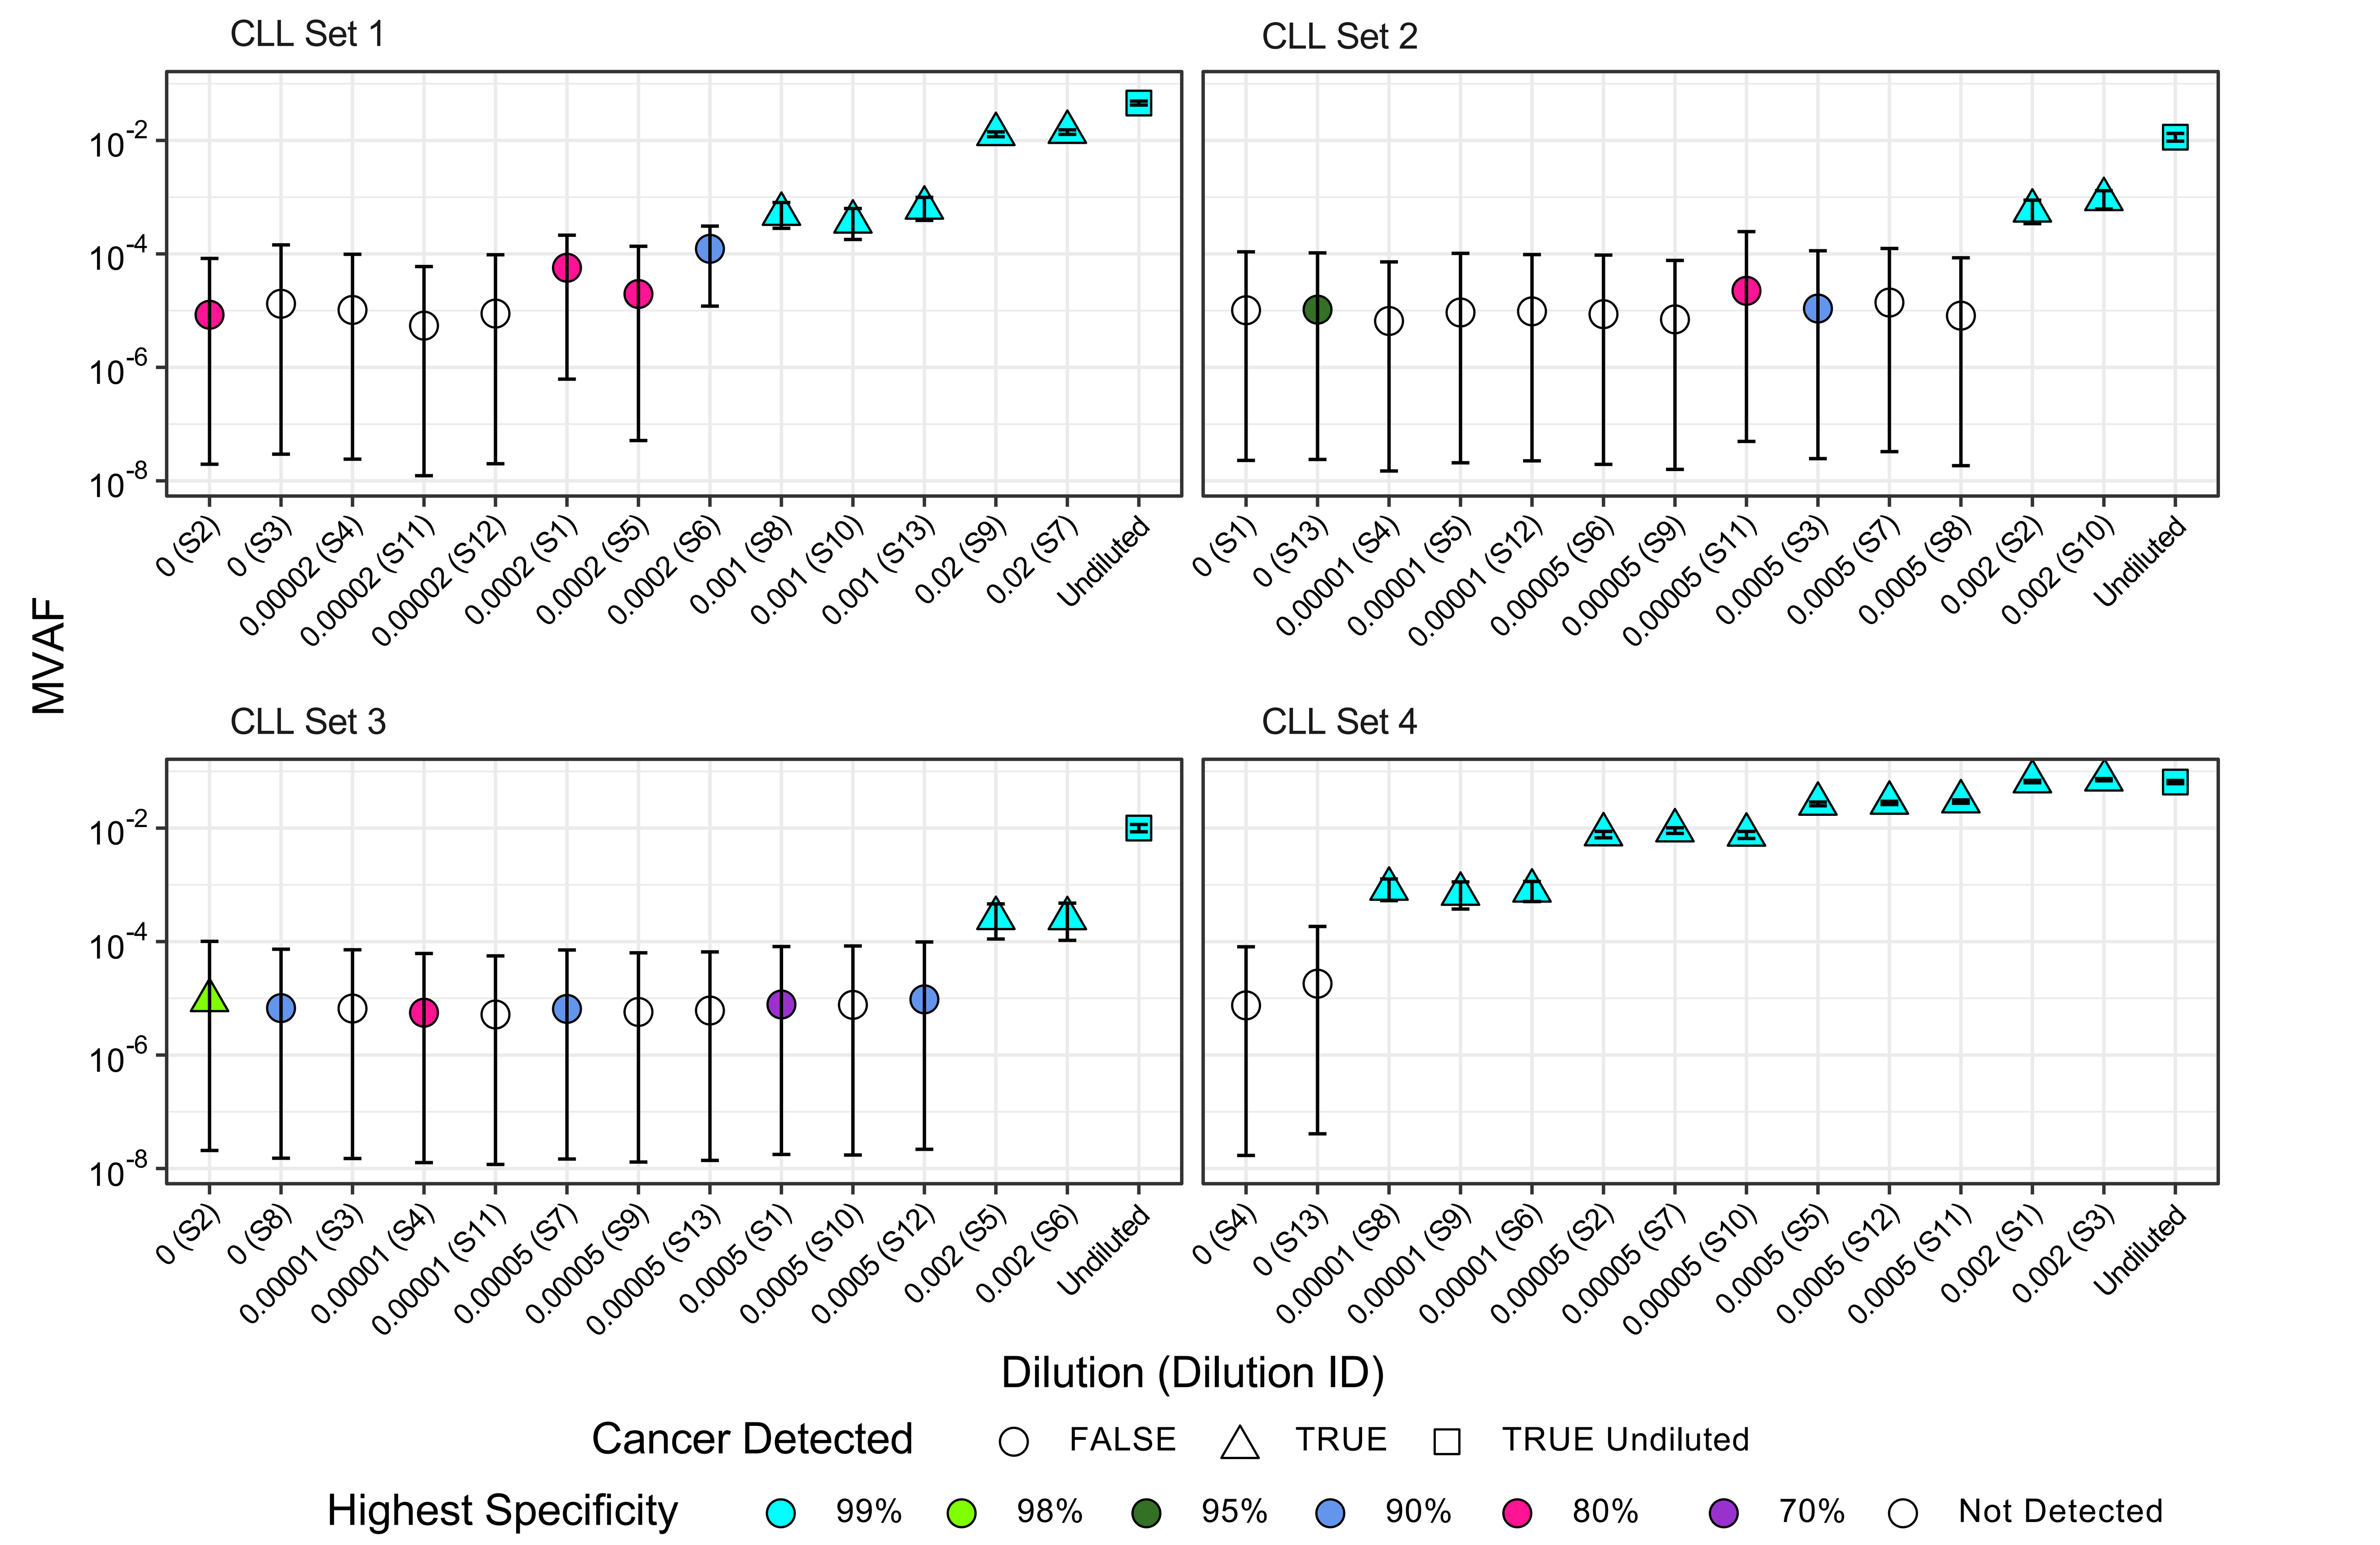

CLL Set 1
CLL Set 2
CLL Set 3
CLL Set 4
MVAF
Dilution (Dilution ID)
Cancer Detected
FALSE
TRUE
TRUE Undiluted
Highest Specificity
99%
98%
95%
90%
80%
70%
Not Detected

Error bars represent 95% CI.